Efficacy and Pharmacokinetics of Cytotoxin Neutralizing Monoclonal Antibodies in Prophylactic and Therapeutic Rabbit Models of *Staphylococcus aureus* Pneumonia

Lukas Stulik[1], Harald Rouha[1], Delphine Labrousse[2], Nelson Anzala[2], Zehra C. Visram[1], Delphine Croisier[2], Gábor Nagy[1], Eszter Nagy[1]

[1] Arsanis Biosciences GmbH, Vienna, Austria, [2] Vivexia Laboratory, Dijon, France

Introduction

Staphylococcus aureus pneumonia is associated with high mortality, irrespective of antibiotic susceptibility. Up to six cytolytic toxins are produced by *S. aureus*: alpha-hemolysin (Hla) and the five bi-component leukocidins (HlgAB, HlgCB, LukED, LukSF, and LukGH/LukAB) that are important for pathogenesis. We previously described two human IgG1 monoclonal antibodies (mAb), ASN-1 and ASN-2, that in combination (as ASN100) neutralize these six toxins. Unlike mice, rabbits are sensitive to all of these toxins and therefore, the prophylactic efficacy and key pharmacokinetic (PK) parameters of ASN100 were determined in this species. Since antibiotics lack anti-toxin mechanisms and are even reported to upregulate toxin production of *S. aureus*, we also evaluated the therapeutic synergy of ASN100 with different antibiotics in a rabbit model of lethal MRSA pneumonia.

Methods

Animals: All studies were performed in ~3kg male New Zealand White rabbits.

Pharmacokinetics: Serum and bronchoalveolar lavage fluid (BALF) samples were serially collected from uninfected and *S. aureus* infected animals immunized with 40 mg/kg of ASN100. Total human and rabbit IgG concentrations were determined by sandwich ELISA using human or rabbit specific capture and detection antibodies. ASN-1 and ASN-2 (free drug) concentrations were measured with anti-idiotype mAbs. Urea levels in BALF and serum were quantified in ELISA and used to determine antibody concentrations in epithelial lining fluid (ELF).

Prophylaxis: Animals were passively immunized with ASN100 at five dose levels between 0.08 to 20 mg/kg total mAb (ASN-1 and ASN-2 at 1:1 ratio), 24 hours prior to intratracheal challenge with lethal doses of USA300 CA-MRSA, USA100 HA-MRSA, or two MSSA strains. Control animals were treated with placebo. Survival was monitored for 7 days post challenge. Alternatively, animals were sacrificed at 12 hours post challenge and gross necropsy as well as microbiological analyses were performed.

Therapy: Animals were intratracheally infected with a lethal dose of a USA300 CA-MRSA strain. Two hours post infection, animals were treated with individually partially protective doses of cloxacillin and/or ASN100. Study drugs were administered at doses and routes stated in the respective panels. Survival of animals was monitored for 7 days post challenge.

Pharmacokinetics of ASN100 in non-infected and infected rabbits

Serum and ELF concentrations of rabbits immunized with 40 mg/kg of ASN100 were determined at different time points after mAb administration. In challenge studies, lethal doses of USA300 CA-MRSA (LAC) or MSSA (ST72) strains were administered intratracheally 24 hours post dosing.

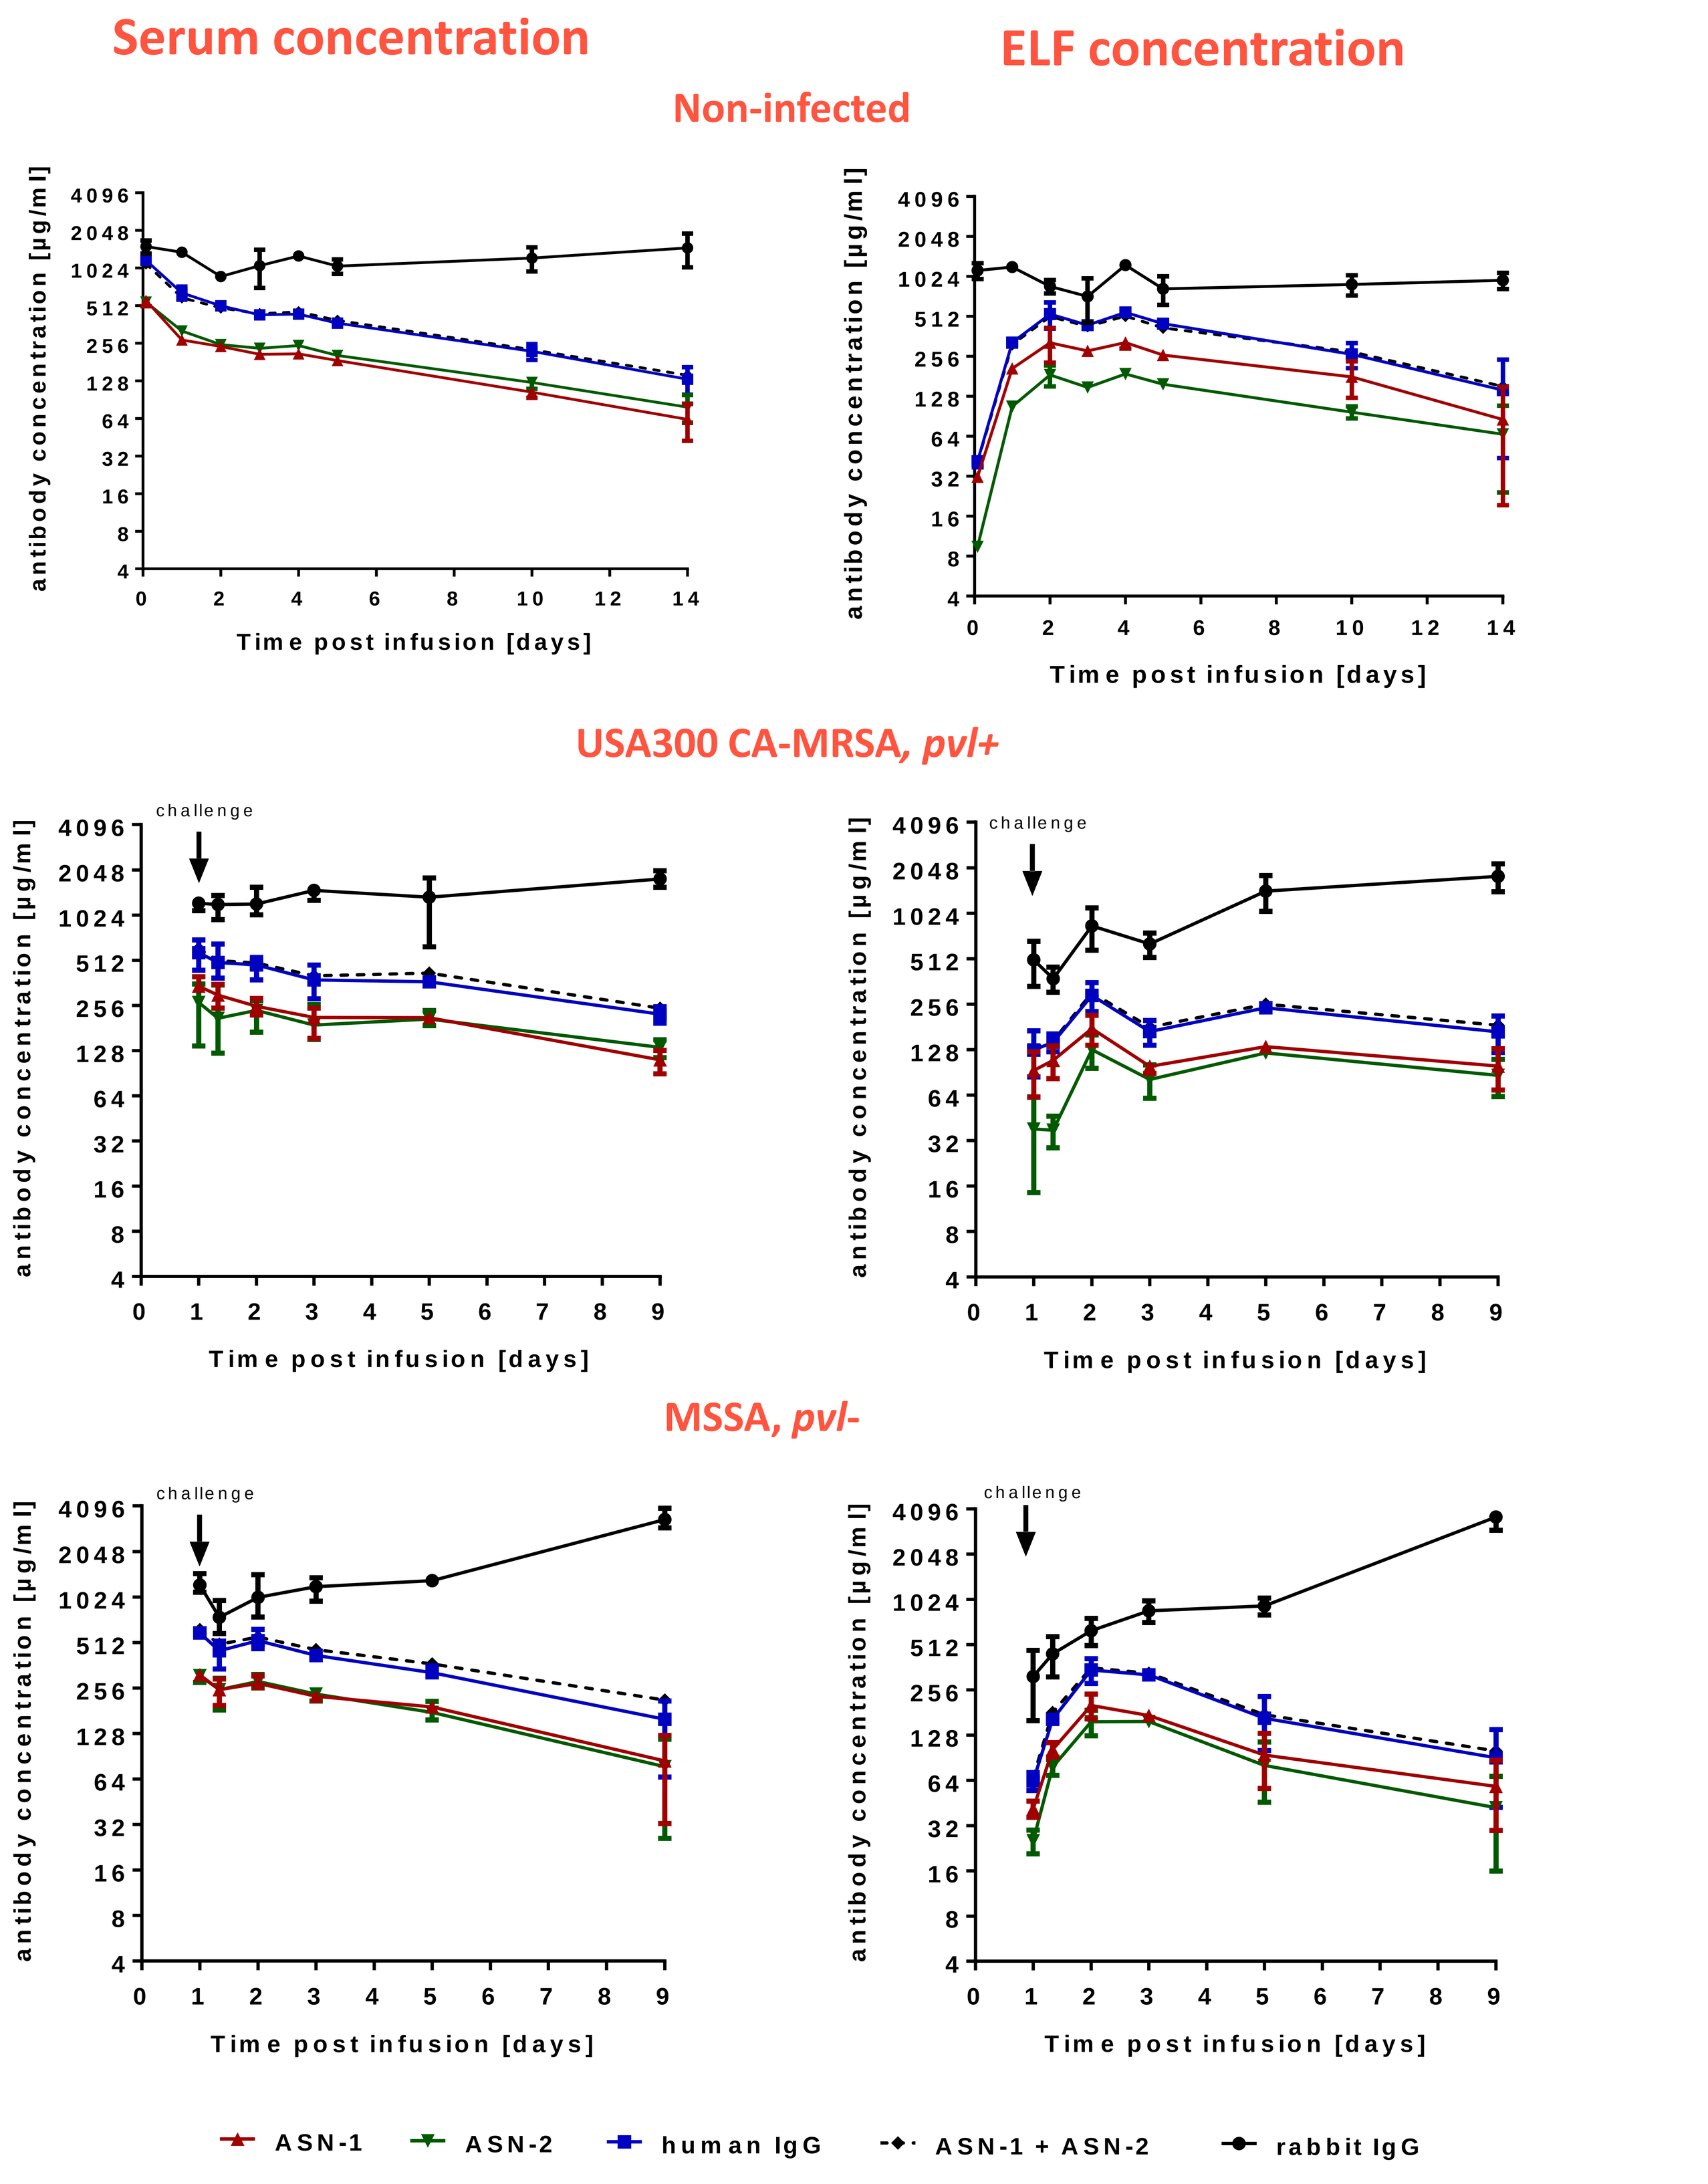
Serum concentration
ELF concentration
Non-infected
USA300 CA-MRSA, pvl+
MSSA, pvl-
challenge
antibody concentration [µg/ml]
Time post infusion [days]
ASN-1
ASN-2
human IgG
ASN-1 + ASN-2
rabbit IgG

Clearance of *S. aureus* from the lungs of ASN100 protected rabbits

Bacterial lung burden was determined in rabbits immunized with 40 mg/kg of ASN100 and challenged with lethal doses of a USA300 CA-MRSA or an MSSA 24 hours after mAb administration.

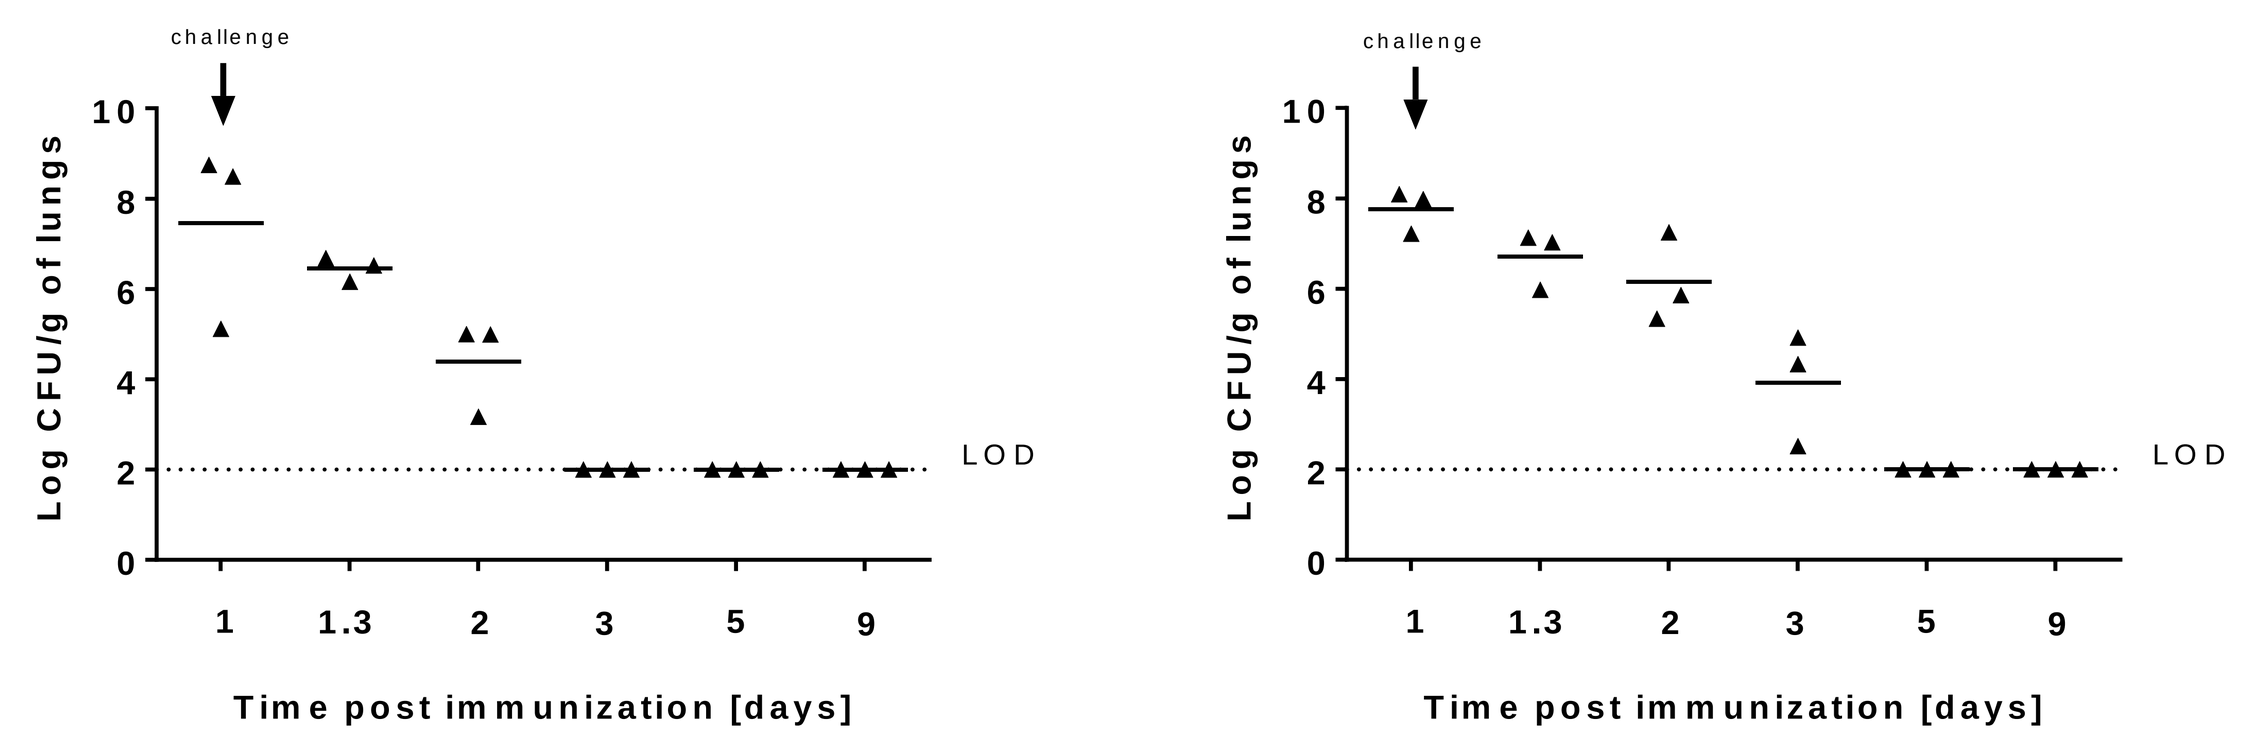
USA300 CA-MRSA, pvl+
MSSA, pvl-
challenge
Log CFU/g of lungs
LOD
Time post immunization [days]

Prophylactic efficacy of in rabbit pneumonia models

24 hours post immunization with different doses of ASN100 or placebo rabbits were intratracheally infected with a lethal incoulum of *S. aureus*. Survival was monitored for 7 days post challenge.

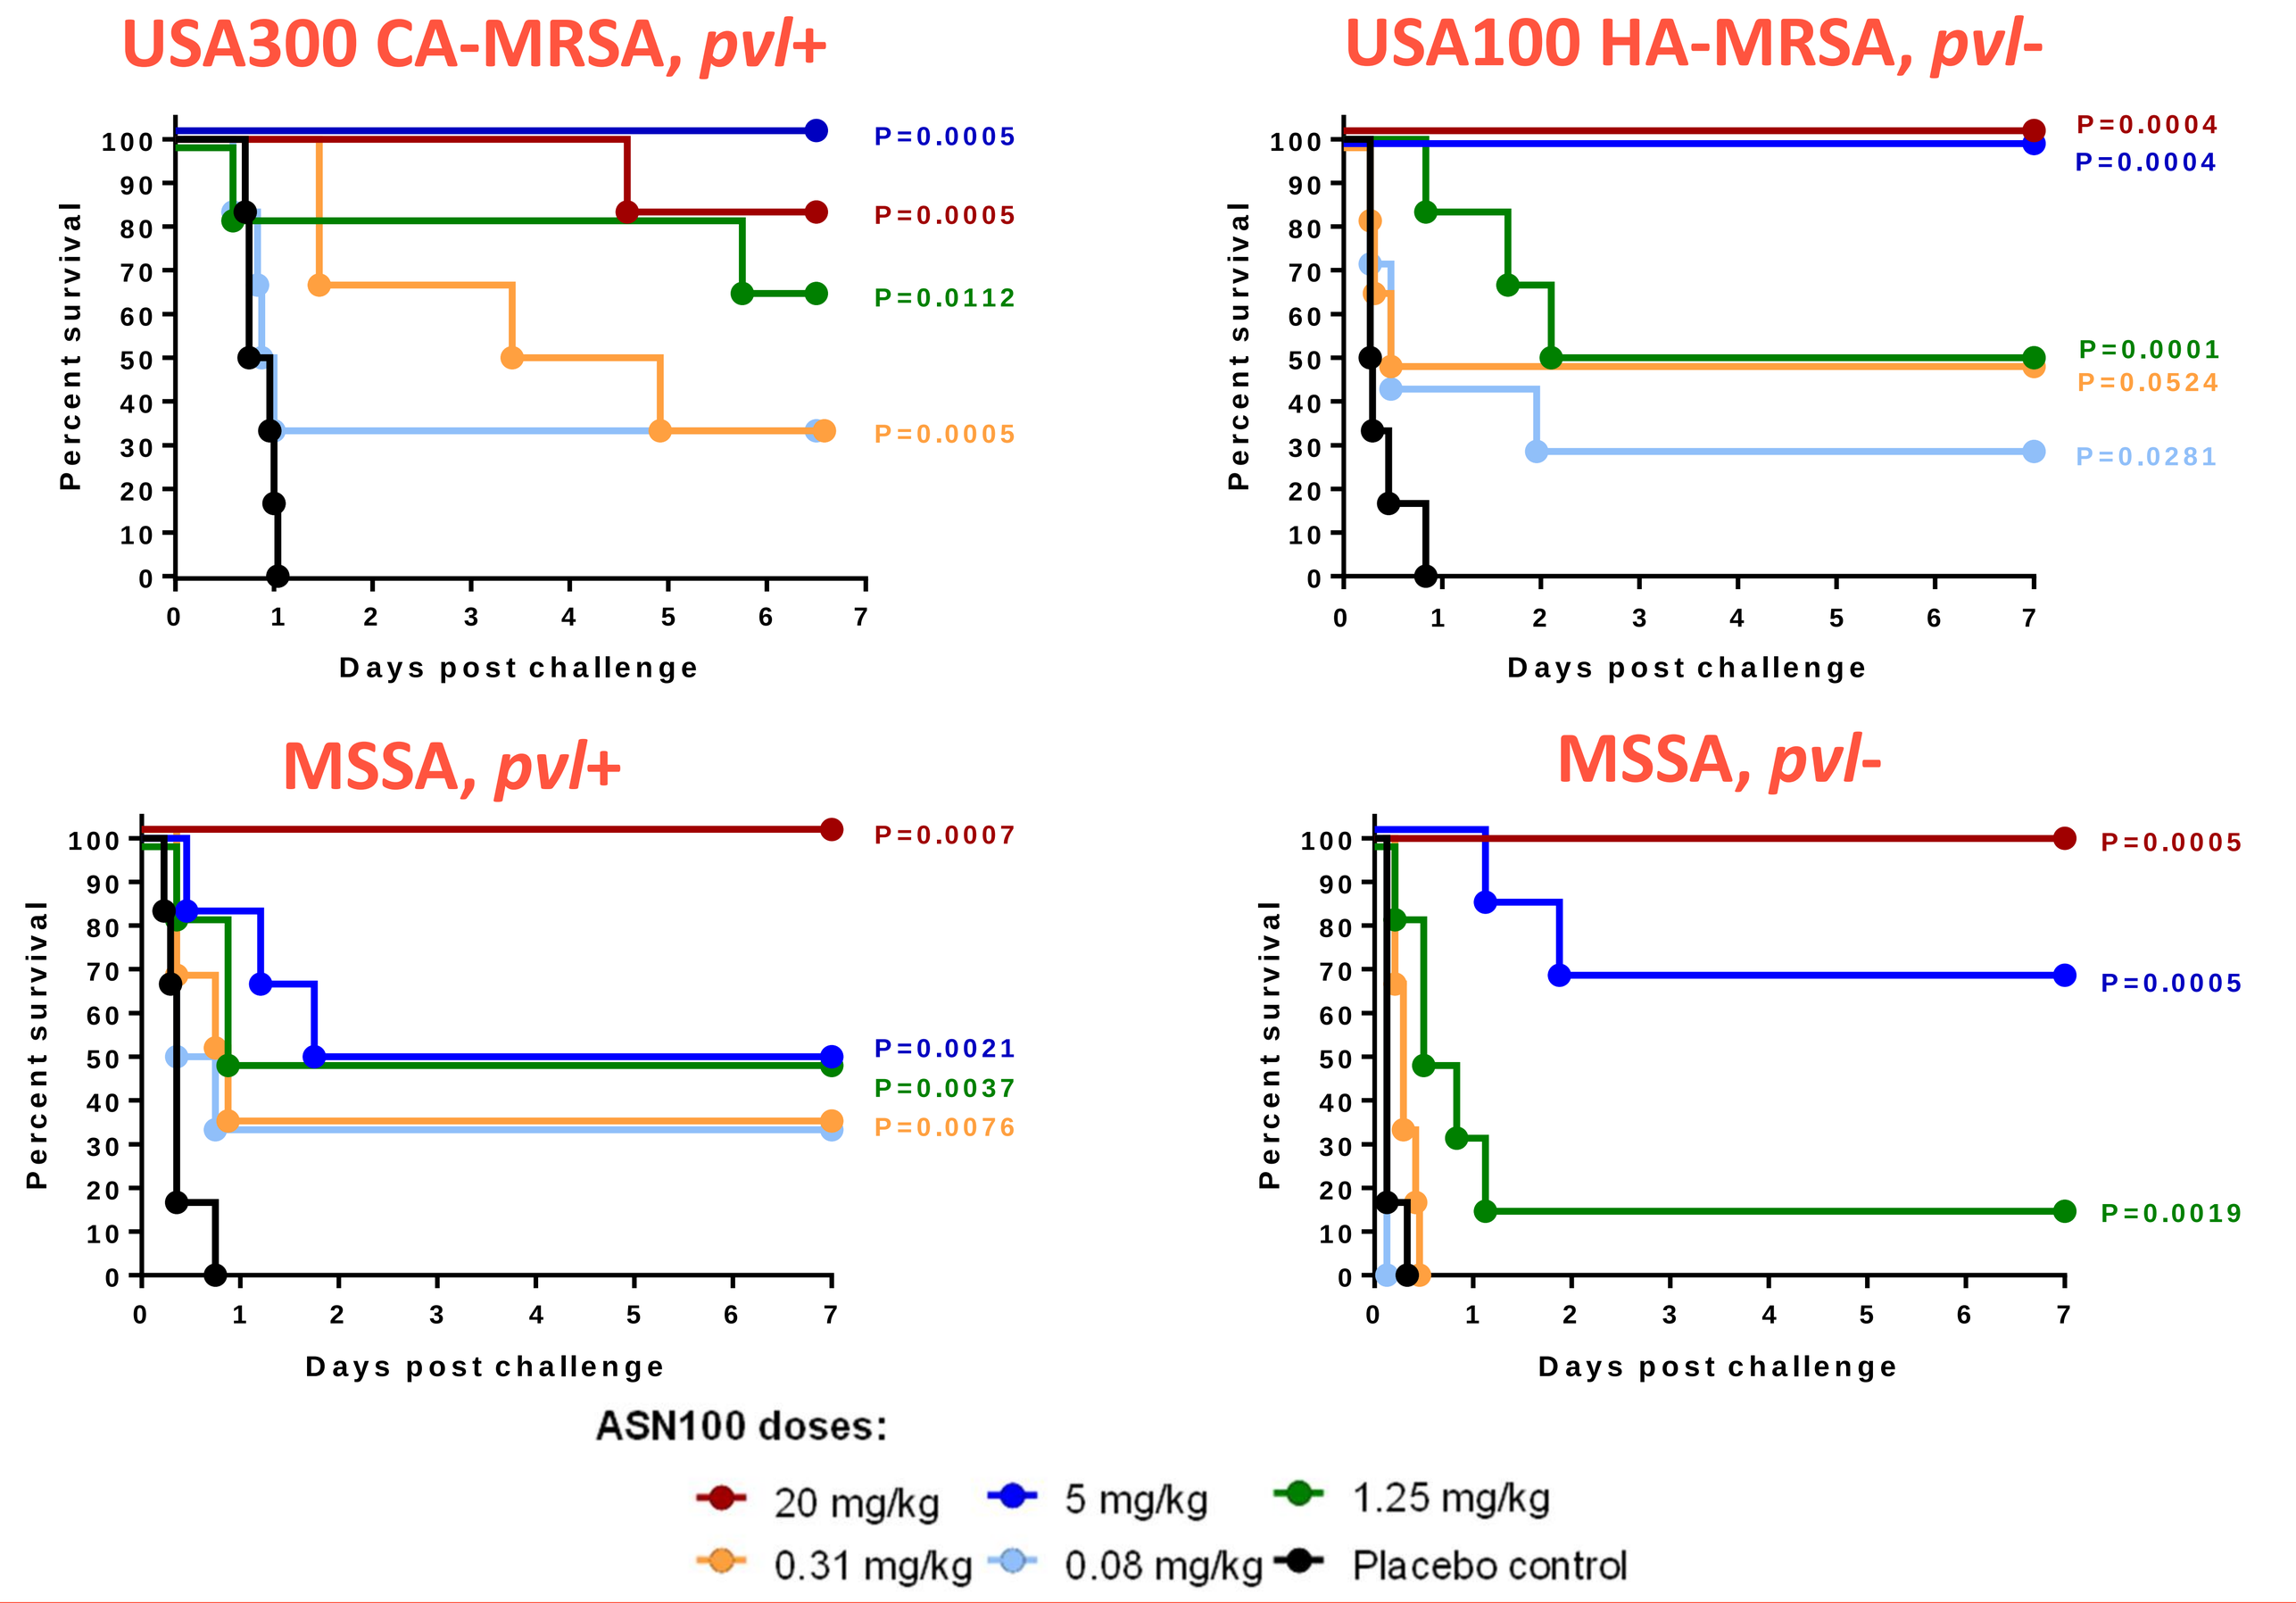
USA300 CA-MRSA, pvl+
P=0.0005
P=0.0005
P=0.0112
P=0.0005
USA100 HA-MRSA, pvl-
P=0.0004
P=0.0004
P=0.0001
P=0.0524
P=0.0281
MSSA, pvl+
P=0.0007
P=0.0021
P=0.0037
P=0.0076
MSSA, pvl-
P=0.0005
P=0.0005
P=0.0019
Percent survival
Days post challenge
ASN100 doses:
20 mg/kg
5 mg/kg
1.25 mg/kg
0.31 mg/kg
0.08 mg/kg
Placebo control

ASN100 dose-dependent effect on lung pathology

Three doses of ASN100 (1.25-20 mg/kg) were evaluated for reduction of lung pathology in infected animals sacrificed 12 hours post-challenge by a USA300 CA-MRSA (LAC) strain.

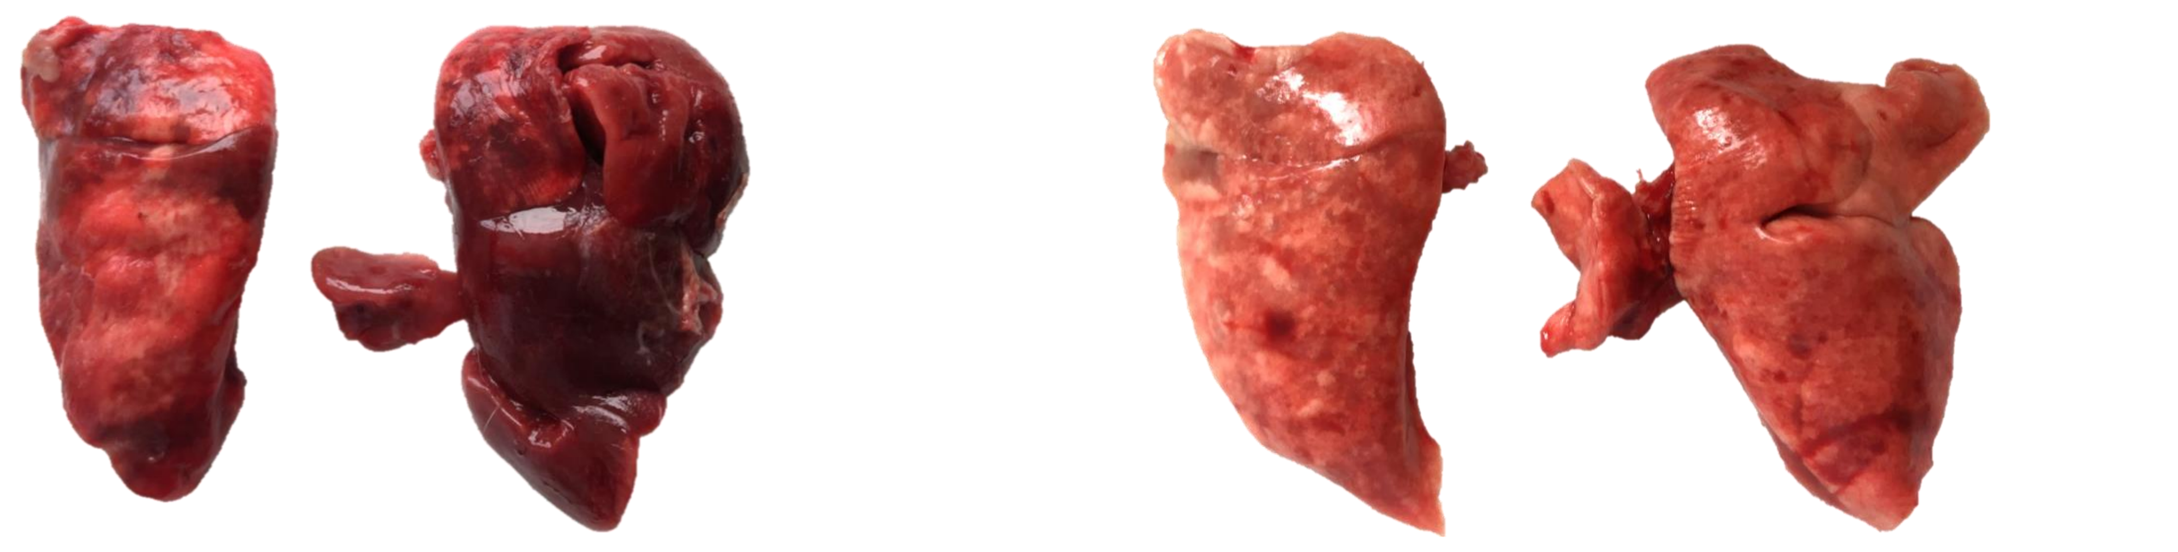
Placebo
ASN100 (20 mg/kg)

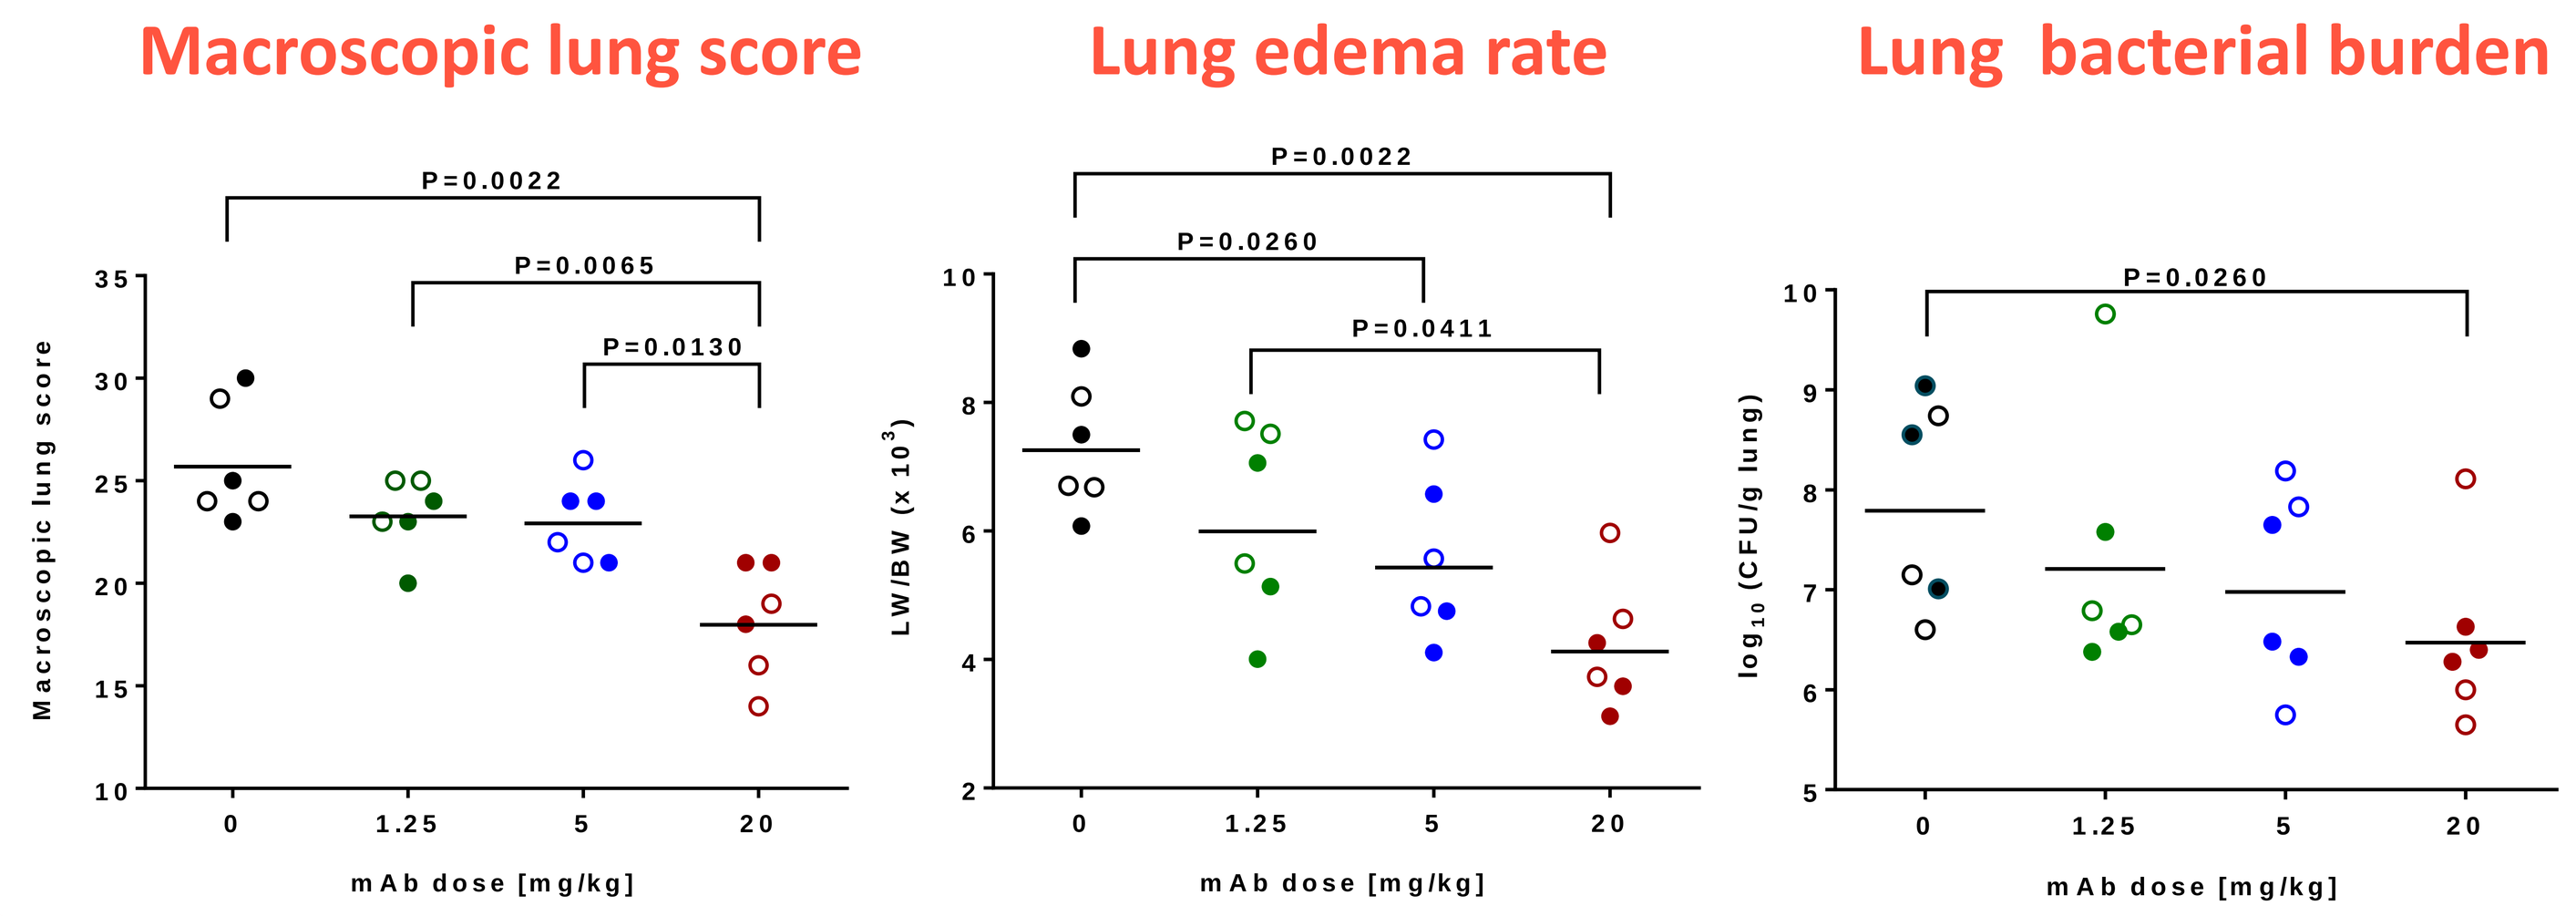
Macroscopic lung score
Lung edema rate
Lung bacterial burden
P=0.0022
P=0.0065
P=0.0130
P=0.0022
P=0.0260
P=0.0411
P=0.0260
Macroscopic lung score
LW/BW (x 10³)
log10 (CFU/g lung)
mAb dose [mg/kg]

ASN100 elicits therapeutic efficacy against a USA300 CA-MRSA in rabbits, which can be improved by cloxacillin

Rabbits were intratracheally infected with a USA300 CA-MRSA strain and treated with ASN100 (single i.v. bolus) and/or cloxacillin (intermitted i.v. infusion q6h for 48h). Survival was monitored for 7 days post challenge.

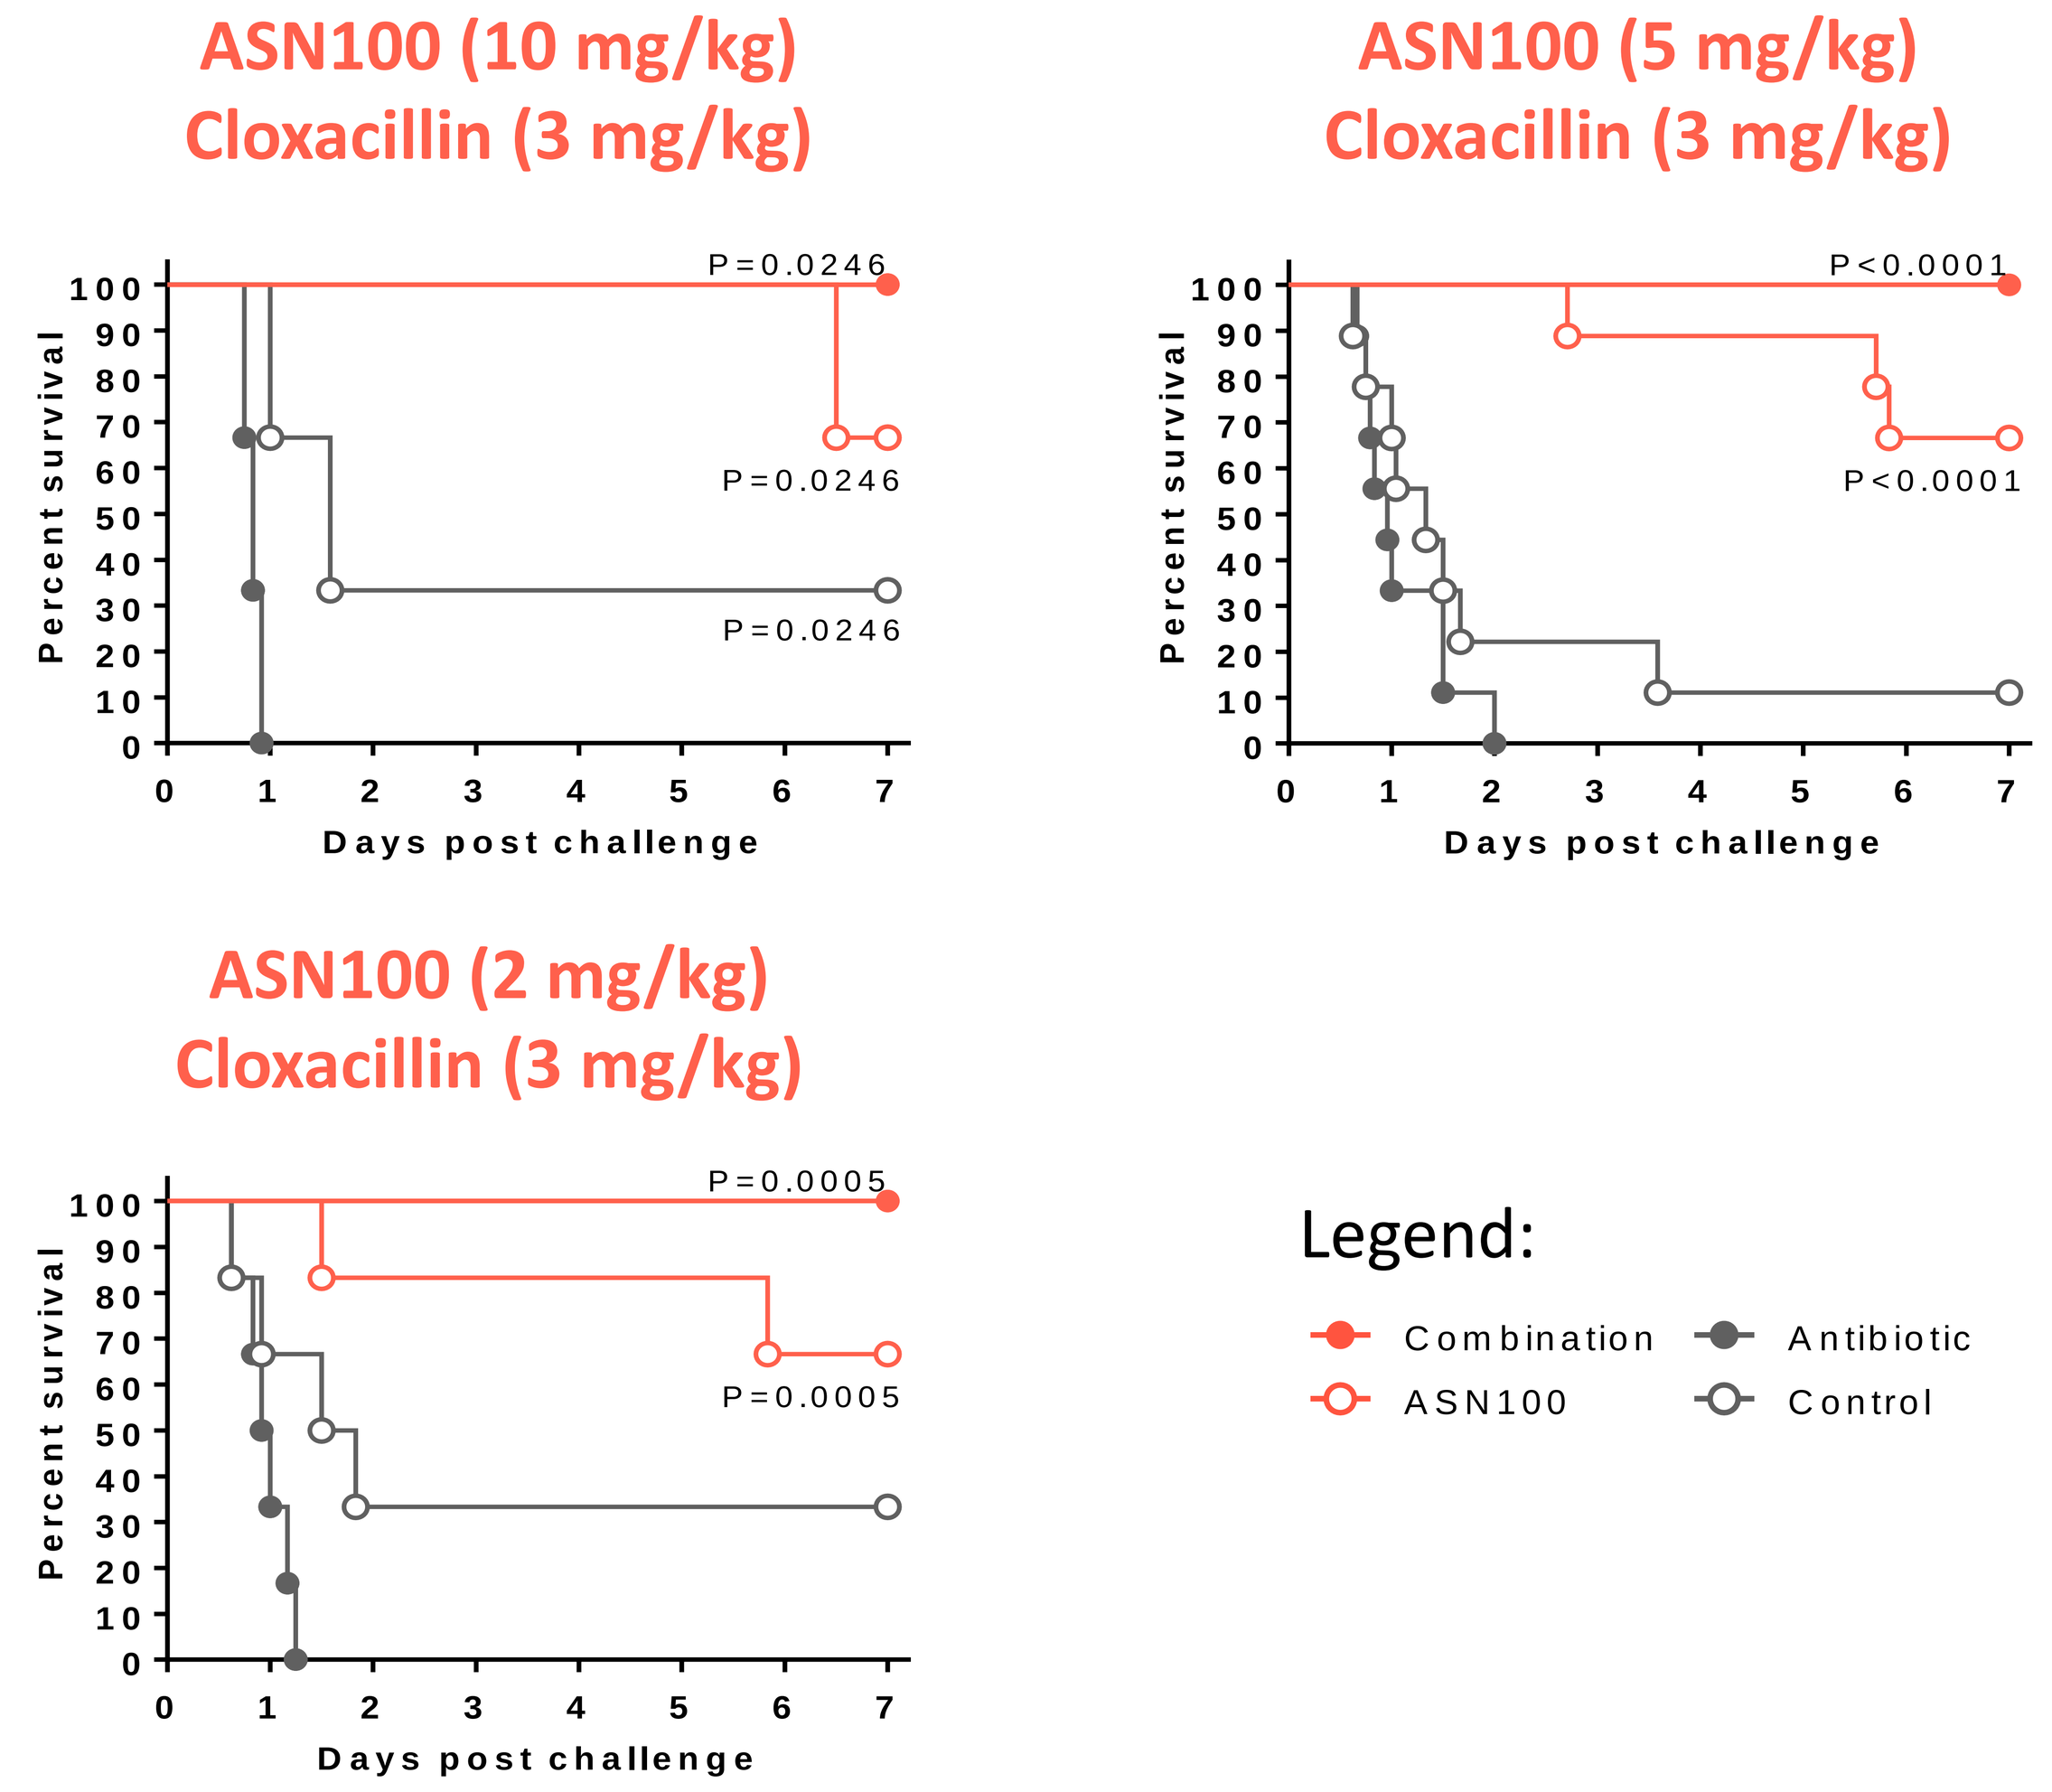
ASN100 (10 mg/kg)
Cloxacillin (3 mg/kg)
P=0.0246
P=0.0246
P=0.0246
ASN100 (5 mg/kg)
Cloxacillin (3 mg/kg)
P<0.0001
P<0.0001
ASN100 (2 mg/kg)
Cloxacillin (3 mg/kg)
P=0.0005
P=0.0005
Percent survival
Days post challenge
Legend:
Combination
Antibiotic
ASN100
Control

Conclusions

- PK-analyses of ASN100 in BALF from uninfected and *S. aureus* infected animals confirmed efficient mAb penetration into lung ELF within the first two days following intravenous administration; ASN100 ELF levels reached 50 % of the serum concentrations by 24 hours, and peak levels by 48 hours post-dosing in uninfected rabbits.
- ASN100 serum and ELF levels were not depleted in the lung of rabbits intratracheally infected with *S. aureus* despite the presence of bacteria for up to 3 days post challenge.
- ASN100 elicited high prophylactic efficacy against all *S. aureus* strains tested in lethal rabbit models of pneumonia irrespective of antibiotic susceptibility (MRSA and MSSA) and toxin expression (both *pvl-* and *pvl+*) profiles of the challenge strains.
- Full protection against the two MRSA strains was achieved by 5 mg/kg ASN100 (2.5 mg/kg each mAb), while the two MSSA strains required a higher dose (20 mg/kg).
- Reduction of bacterial counts, lung edema rate as well as macroscopic lung scores of infected animals were most pronounced with the 20 mg/kg ASN100 dose.
- In therapeutic models, ASN100 showed significant efficacy alone, with doses as low as 2 mg/kg protecting 67% of the rabbits.
- Surprisingly, the combination of ASN100 with cloxacillin further improved the therapeutic efficacy, even against the USA300 CA-MRSA strain.
- ASN100 is currently being evaluated in a Phase 2 clinical trial to prevent *S. aureus* pneumonia in mechanically ventilated patients.

Acknowledgements: We thank Sandrine Albac, Davy Hayez, Karin Groß, Barbara Maierhofer and Susanne Weber for their technical support.

Disclosure: All authors are employees and shareholders in the companies performing this work.

Contact: Lukas Stulik, Arsanis Biosciences GmbH, Helmut-Qualtinger-Gasse 2, 1030 Vienna, Austria. E-mail: lukas.stulik@arsanis.com